A TRAUMATIC TALE OF TWO CITIES, PART ONE:
Does being treated by different EMS affect outcomes in trauma patients destined for transport to level one centres in Halifax and Saint John?

Colin Rouse[1], Jefferson Hayre[1], Dr. James French[2], Ian Watson[3], Allison Chisholm[3], Sue Benjamin[3], Jacqueline Fraser[4], Beth Sealy[5], Dr. Mete Erdogan[5], Dr. Robert Green[5,6], Dr. Paul Atkinson[1,2,3]

1. Dalhousie Medicine New Brunswick, Saint John, New Brunswick
2. Department of Emergency Medicine, Dalhousie University, Saint John Regional Hospital, Saint John, New Brunswick
3. New Brunswick Trauma Program, Saint John, New Brunswick
4. Department of Emergency Medicine, Saint John Regional Hospital, Saint John, New Brunswick
5. Nova Scotia Trauma Program, Halifax, Nova Scotia
6. Departments of Emergency Medicine and Clinical Care Medicine, Dalhousie University, QEII Health Sciences Centre, Halifax, Nova Scotia

CONTE DE TRAUMATOLOGIE DE DEUX VILLES, PREMIÈRE PARTIE :
Les traitements donnés par différents services médicaux d'urgence influencent-ils les résultats chez les patients qui reçoivent des soins de traumatologie pendant leur transport vers un centre de traumatologie de niveau un de Halifax et de Saint John?

Colin Rouse[1], Jefferson Hayre[1], Dr James French[2], Ian Watson[3], Allison Chisholm[3], Sue Benjamin[3], Jacqueline Fraser[4], Beth Sealy[5], Dr Mete Erdogan[5], Dr Robert Green[5,6], Dr Paul Atkinson[1,2,3]

1. Dalhousie Medicine New Brunswick, Saint John (Nouveau-Brunswick)
2. Service de médecine d'urgence, Université Dalhousie, Hôpital régional de Saint John, Saint John (Nouveau-Brunswick)
3. Programme de traumatologie du Nouveau-Brunswick, Saint John (Nouveau-Brunswick)
4. Service de médecine d'urgence, Hôpital régional de Saint John, Saint John (Nouveau-Brunswick)
5. Programme de traumatologie de Nouvelle-Écosse, Halifax (Nouvelle-Écosse)
6. Service de médecine d'urgence et de médecine des soins cliniques, Université Dalhousie, QEII Health Sciences Centre, Halifax (Nouvelle-Écosse)

Introduction

Advanced Emergency Medical Systems (AEMS) have been implemented in various regions worldwide despite the fact that prior evidence has not established a conclusive survival benefit when compared to Standard Emergency Medical Systems (SEMS). The presence of an AEMS in Nova Scotia (NS) and a SEMS in New Brunswick (NB) provides a natural experiment where two different systems deliver services to populations with similar demographics in Atlantic Canada.

Objective

This study sought to determine if there were differences in overall patient survival rates, survival rates of severely injured patients (ISS>24), and the interventions performed between an AEMS in NS and a SEMS in NB.

Methods

- Prospective observational cohort study
- April 1, 2011 – March 31, 2013
- 101 cases in NB vs. 251 cases in NS
- Survival to hospital and survival to discharge or thirty days were the primary endpoints
- Hypothesis testing was conducted using Fisher's exact test to compare the results data extracted from the two registries

Patients:

- Age >15
- ISS > 12
- Direct Transport to Level 1 Trauma Centre

Results

Baseline demographics were similar between groups. Overall, there was no difference in survival to hospital when patients were treated by an AEMS (92%, n=232) compared to patients treated by a SEMS (95%, n=96) respectively (p=0.49). Furthermore, when comparing patients with more severe injuries (ISS>24) there was no significant difference in survival. When treated in an AEMS 55% (n=44) of severely injured patients survived to hospital compared to 55% (n=18) of severely injured patients treated in a SEMS (p=1.00)

Differences were observed in the percentages of trauma patients who received interventions in the AEMS compared to those in the SEMS. In the AEMS 15% (n=38) of patients received a prehospital airway intervention compared to 3% (n=3) of patients in the SEMS (p<0.01). There was also a higher number of patients who received prehospital intravenous access in the AEMS (78%, n=195) when compared to the SEMS (31%, n =32; p<0.01). There was no significant difference in the administration of oxygen between the AEMS (66%, n=165) and the SEMS (72%, n=71; p=0.45).

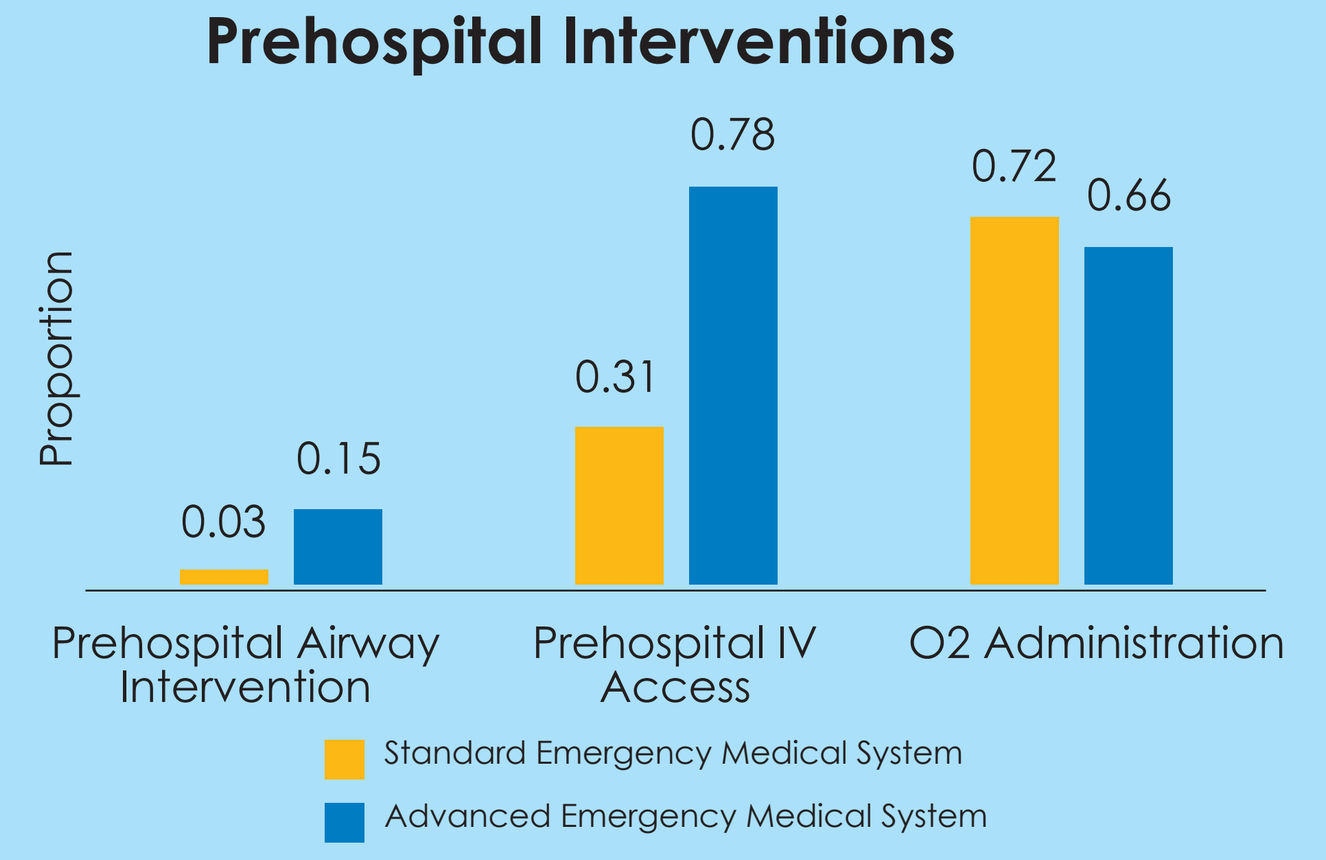

Prehospital Interventions
Proportion
0.03
0.15
0.31
0.78
0.72
0.66
Prehospital Airway Intervention
Prehospital IV Access
O2 Administration
Standard Emergency Medical System
Advanced Emergency Medical System

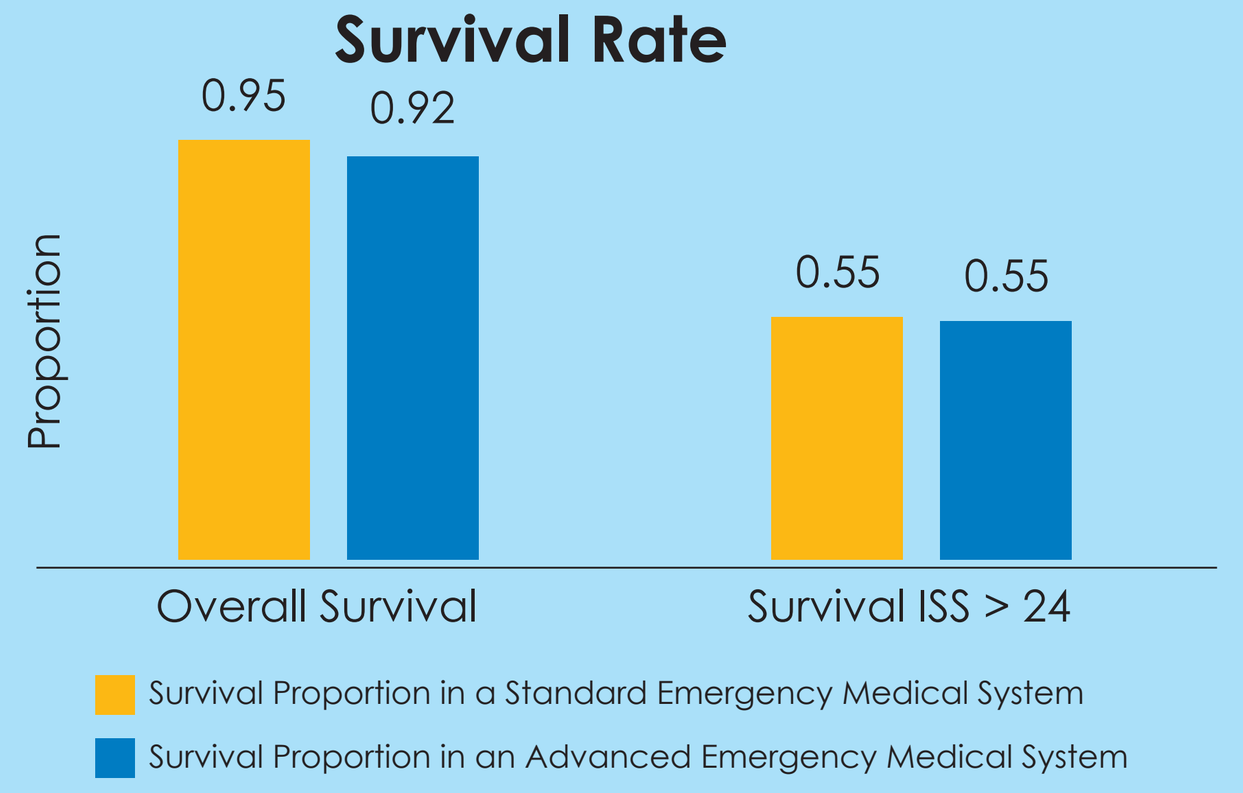

Survival Rate
Proportion
0.95
0.92
0.55
0.55
Overall Survival
Survival ISS > 24
Survival Proportion in a Standard Emergency Medical System
Survival Proportion in an Advanced Emergency Medical System

	SEMS	AEMS	Significance
Overall Survival	0.95 (n=96)	0.92 (n=232)	*p = 0.49*
Survival ISS >24	0.55 (n=18)	0.55 (n=44)	*p = 1.00*
Prehospital Airway Interventions	0.03 (n=3)	0.15 (n=38)	***p < 0.01***
Prehospital IV Access	0.31 (n=32)	0.78 (n=195)	***p < 0.01***
Prehospital O_2 Administration	0.72 (n=71)	0.66 (n=165)	*p = 0.45*

Conclusion

Overall survival to hospital was the same between advanced and standard Canadian EMS systems despite the variability of the interventions performed in each system. As numbers included are low, individual case benefit cannot be excluded. These results support the need for case level data sharing between provinces to permit analysis of potential confounding variables.

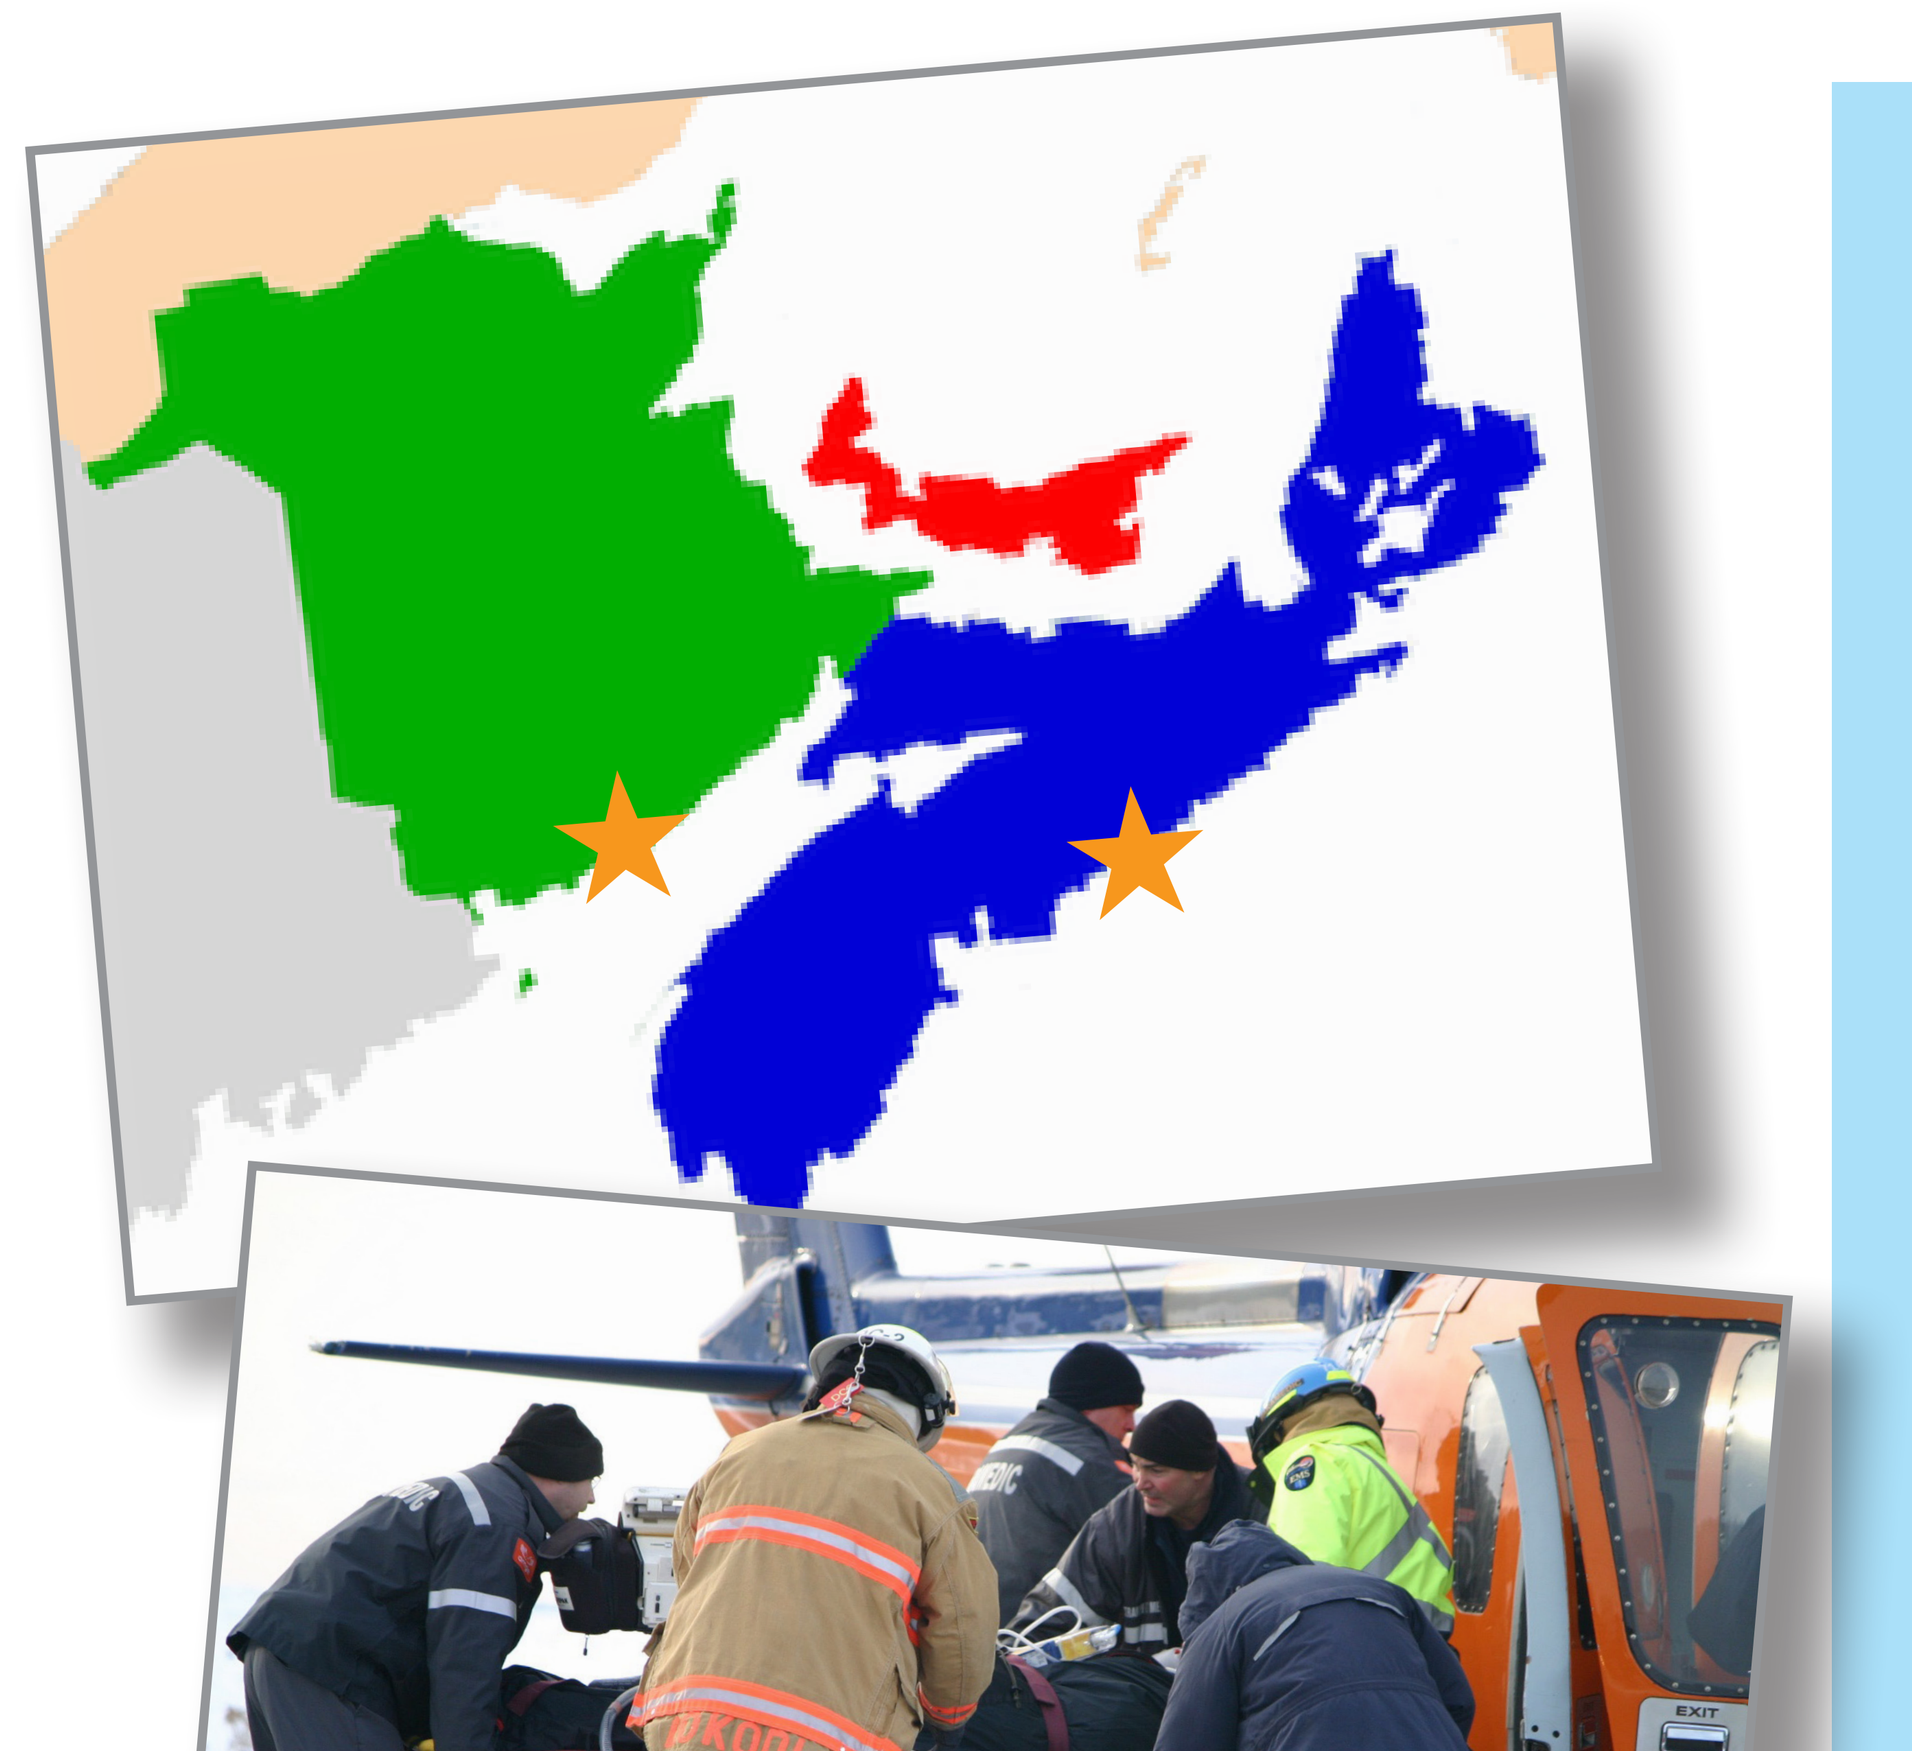

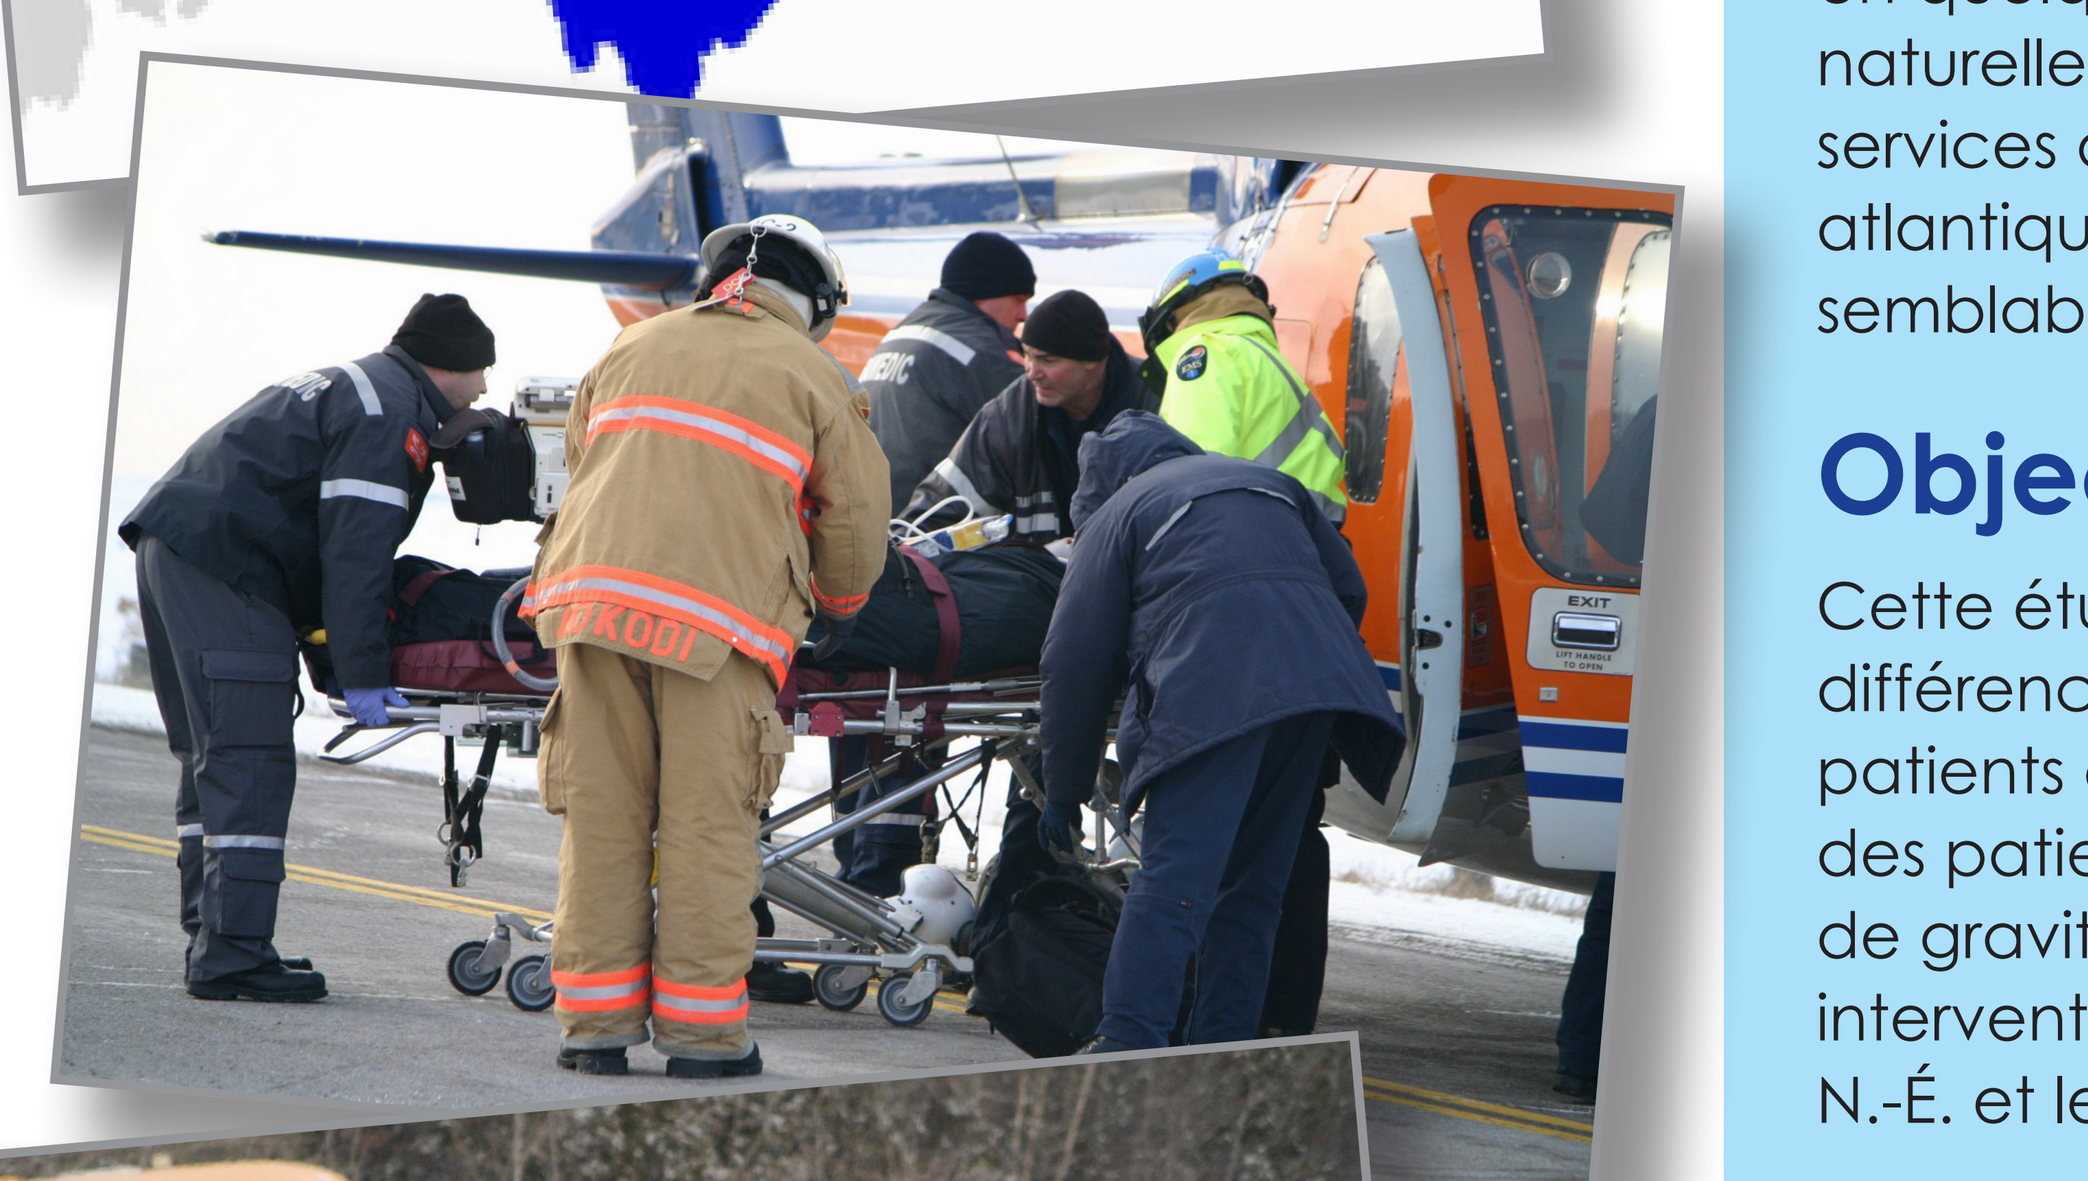

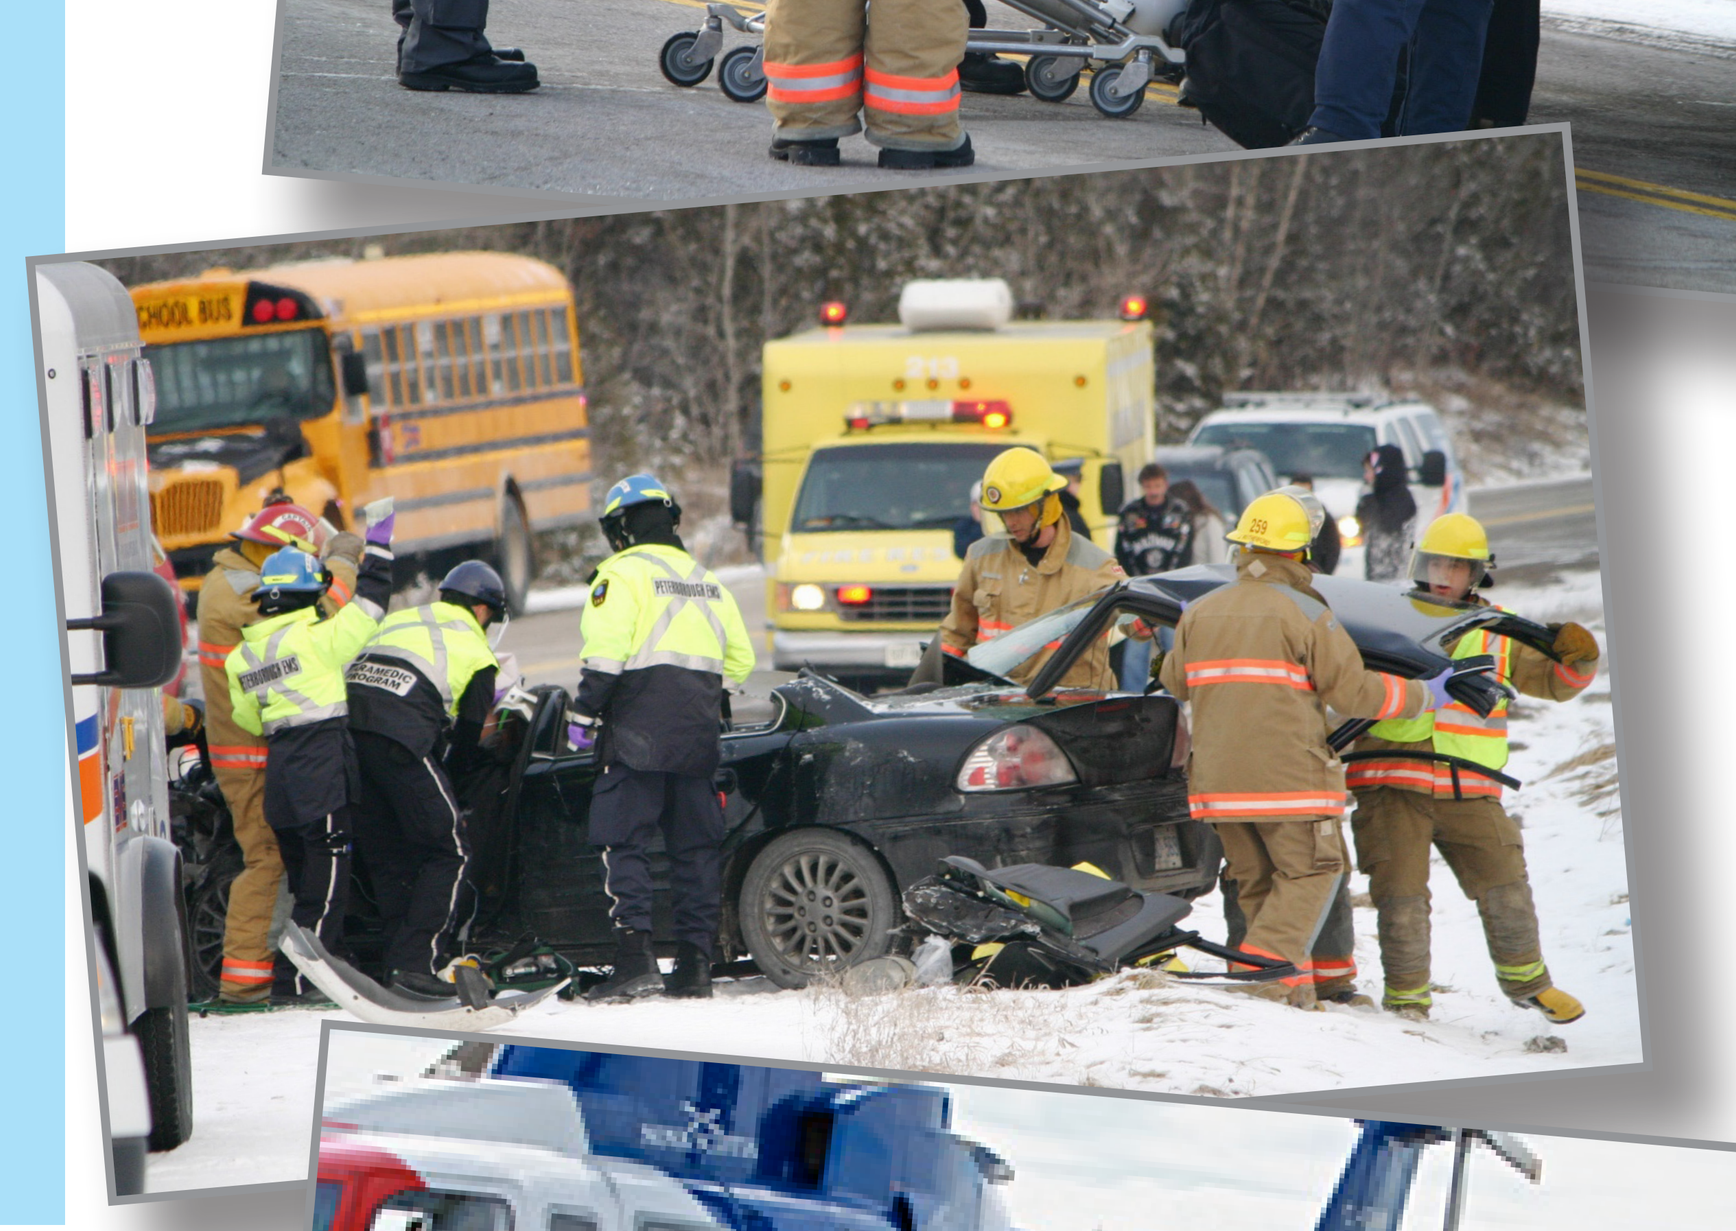

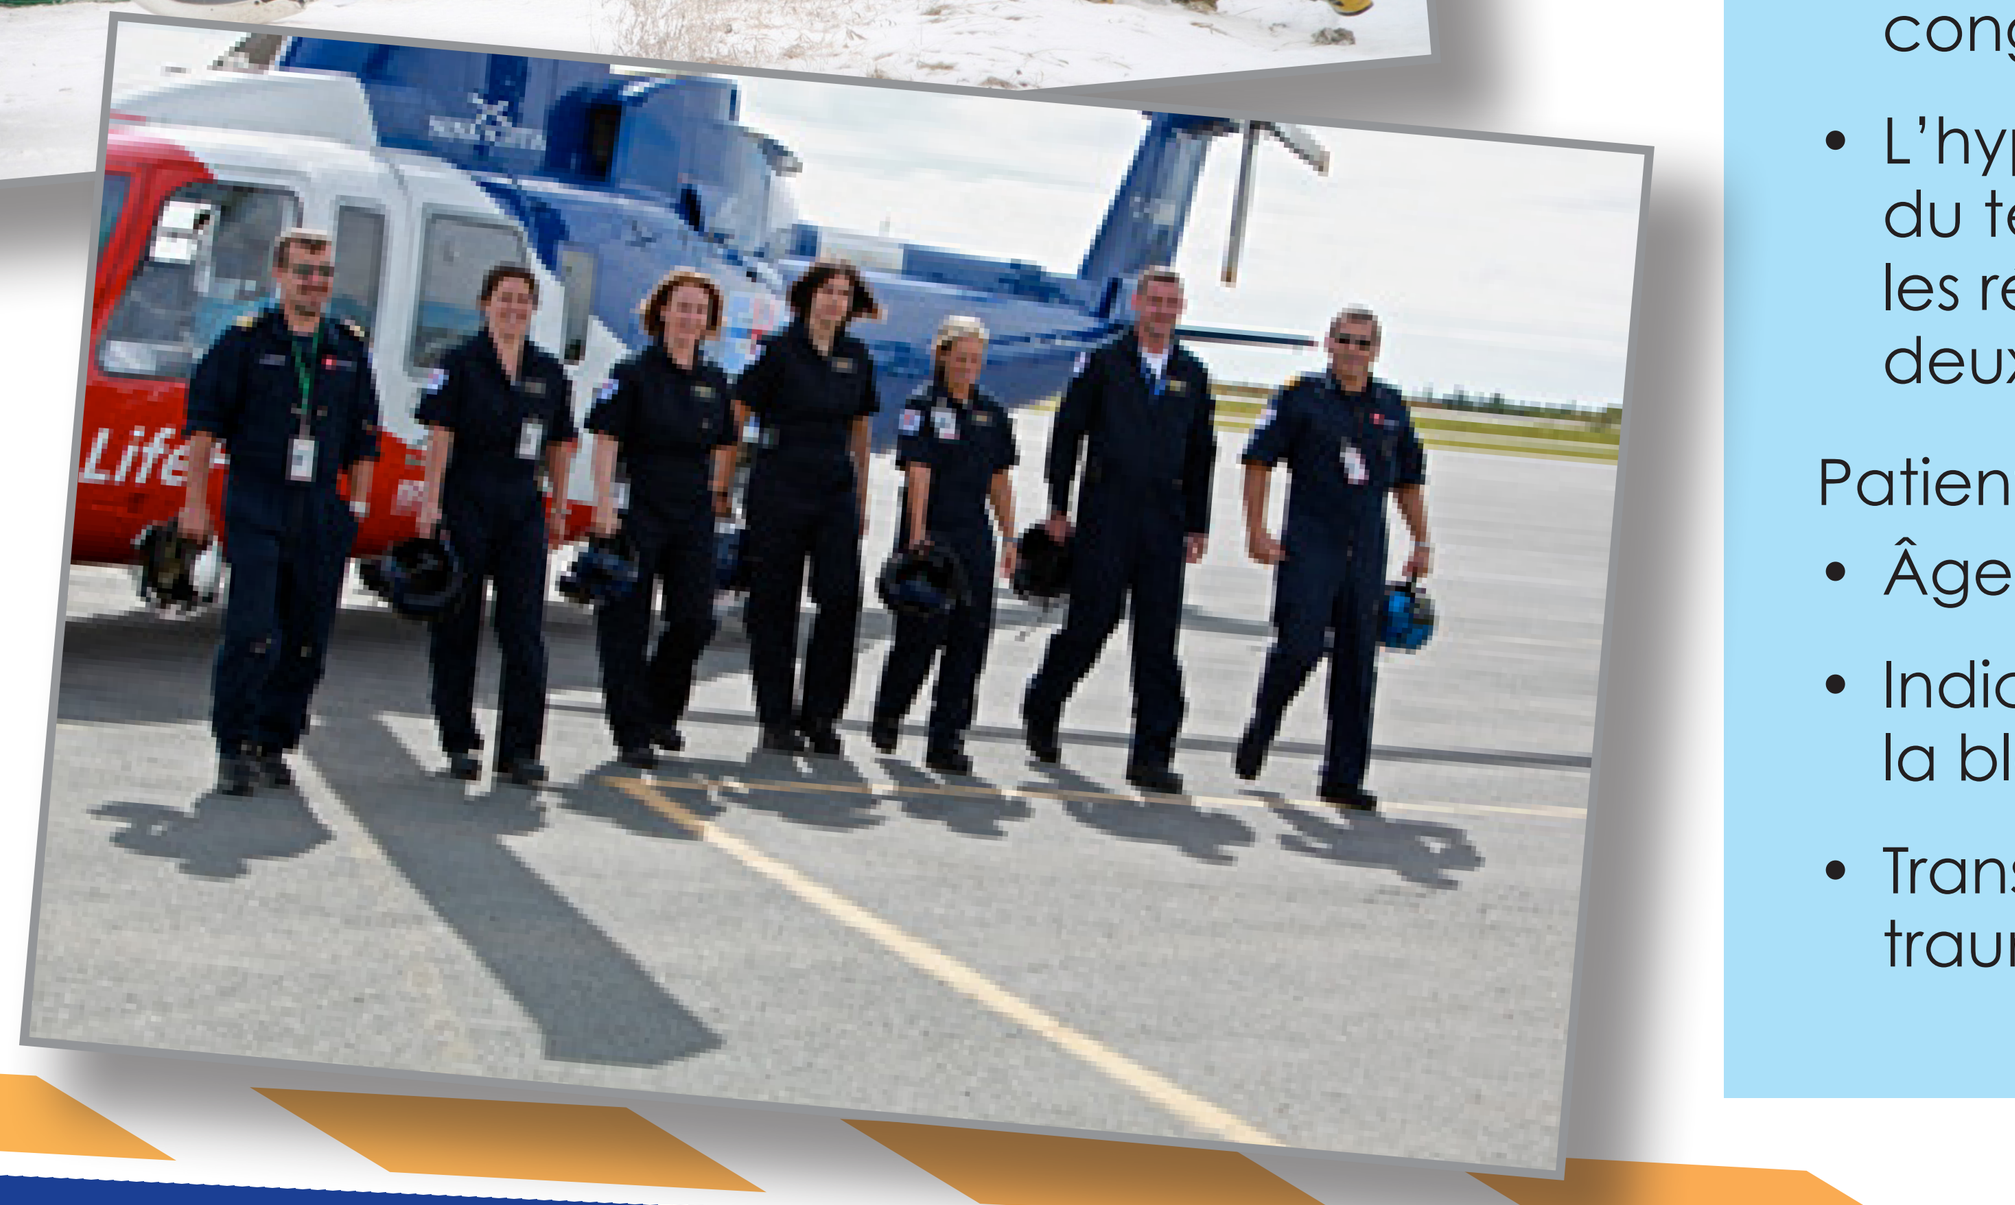

Introduction

Diverses régions du monde ont mis en œuvre des systèmes médicaux d'urgence avancés (SMUA) malgré le fait que les preuves recueillies ne montrent aucun avantage sur le plan de la survie comparativement aux systèmes médicaux d'urgence normalisés (SMUN). Le SMUA en Nouvelle-Écosse (N.-É.) et le SMUN au Nouveau-Brunswick (N.-B.) fournissent en quelque sorte une expérience naturelle où deux systèmes offrent des services à deux populations du Canada atlantique aux données démographiques semblables.

Objectif

Cette étude cherche à déterminer les différences entre les taux de survie des patients en général, les taux de survie des patients grièvement blessés (indice de gravité de la blessure > 24), et les interventions réalisées par le SMUA de la N.-É. et le SMUN du N.-B.

Méthodes

- Étude d'observation prospective de cohorte
- Du 1er avril 2011 au 31 mars 2013
- 101 cas au N.-B. et 251 cas en N.-É.
- Principaux indicateurs de résultats : taux de survie jusqu'à l'hôpital et jusqu'au congé de l'hôpital ou après 30 jours
- L'hypothèse a été vérifiée au moyen du test exact de Fisher pour comparer les résultats des données recueillies des deux registres

Patients :

- Âge > 15
- Indice de gravité de la blessure (IGB) > 12
- Transport direct au centre de traumatologie de niveau 1

Results

Les données démographiques de référence des deux groupes étaient semblables. En général, il n'y avait pas de différence entre le taux de survie à l'hôpital lorsqu'on traitait les patients dans un SMUA (92 %, n = 232) et lorsqu'on les traite dans un SMUN (95 %, n = 96) respectivement (*p* = 0,49). De plus, il n'y a pas de différence importante du taux de survie des patients ayant des blessures plus graves (IGB > 24). Lorsqu'ils étaient traités dans un SMUA, 55 % (n = 44) des patients grièvement blessés ont survis jusqu'à l'hôpital contre 55 % (n = 18) des patients traités dans un SMUN (*p* = 1,00).

On a observé quelques différences dans le pourcentage des patients victimes d'un traumatisme qui ont reçu des interventions dans le SMUA comparativement à celui dans le SMUN. Dans le SMUA, 15 % (n = 38) des patients ont reçu une intervention préhospitalière sur les voies respiratoires contre 3 % (n = 3) des patients dans le SMUN (*p* < 0,01). Le nombre de patients ayant reçu un accès intraveineux avant l'arrivée à l'hôpital dans le SMUA (78 %, n = 195) est plus élevé que celui dans le SMUN (31 %, n = 32; *p* < 0,01). Aucune différence importante n'a été observée entre l'administration d'oxygène dans le SMUA (66 %, n = 165) et dans le SMUN (72 %, n = 71; *p* = 0,45).

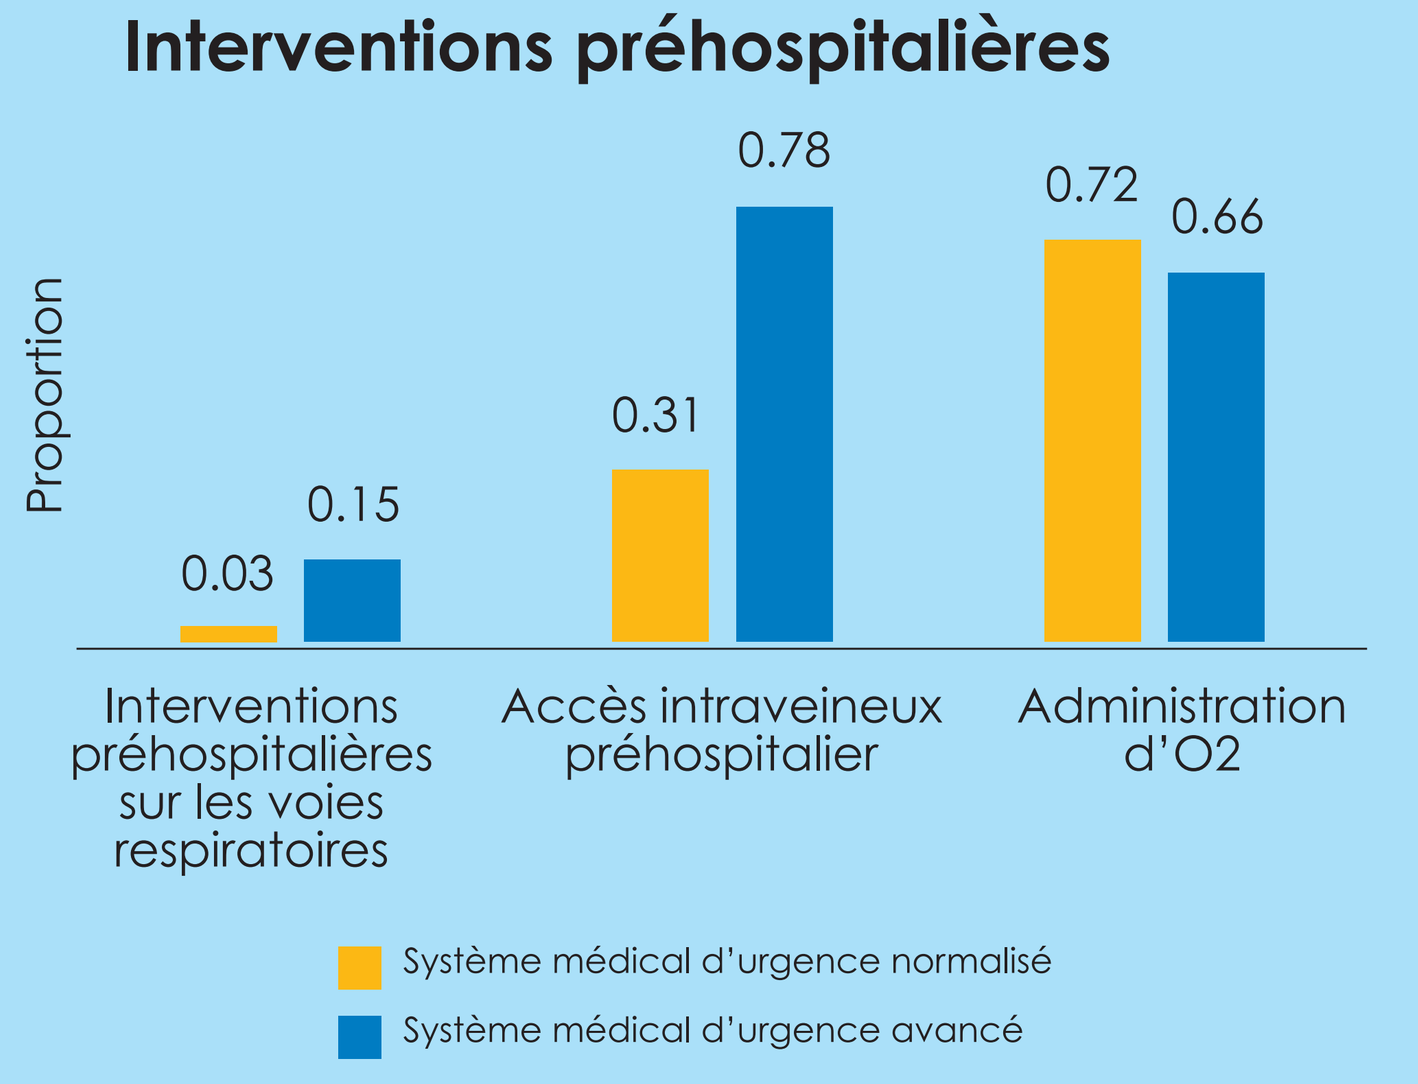

Interventions préhospitalières
Proportion
0.03
0.15
0.31
0.78
0.72
0.66
Interventions préhospitalières sur les voies respiratoires
Accès intraveineux préhospitalier
Administration d'O2
Système médical d'urgence normalisé
Système médical d'urgence avancé

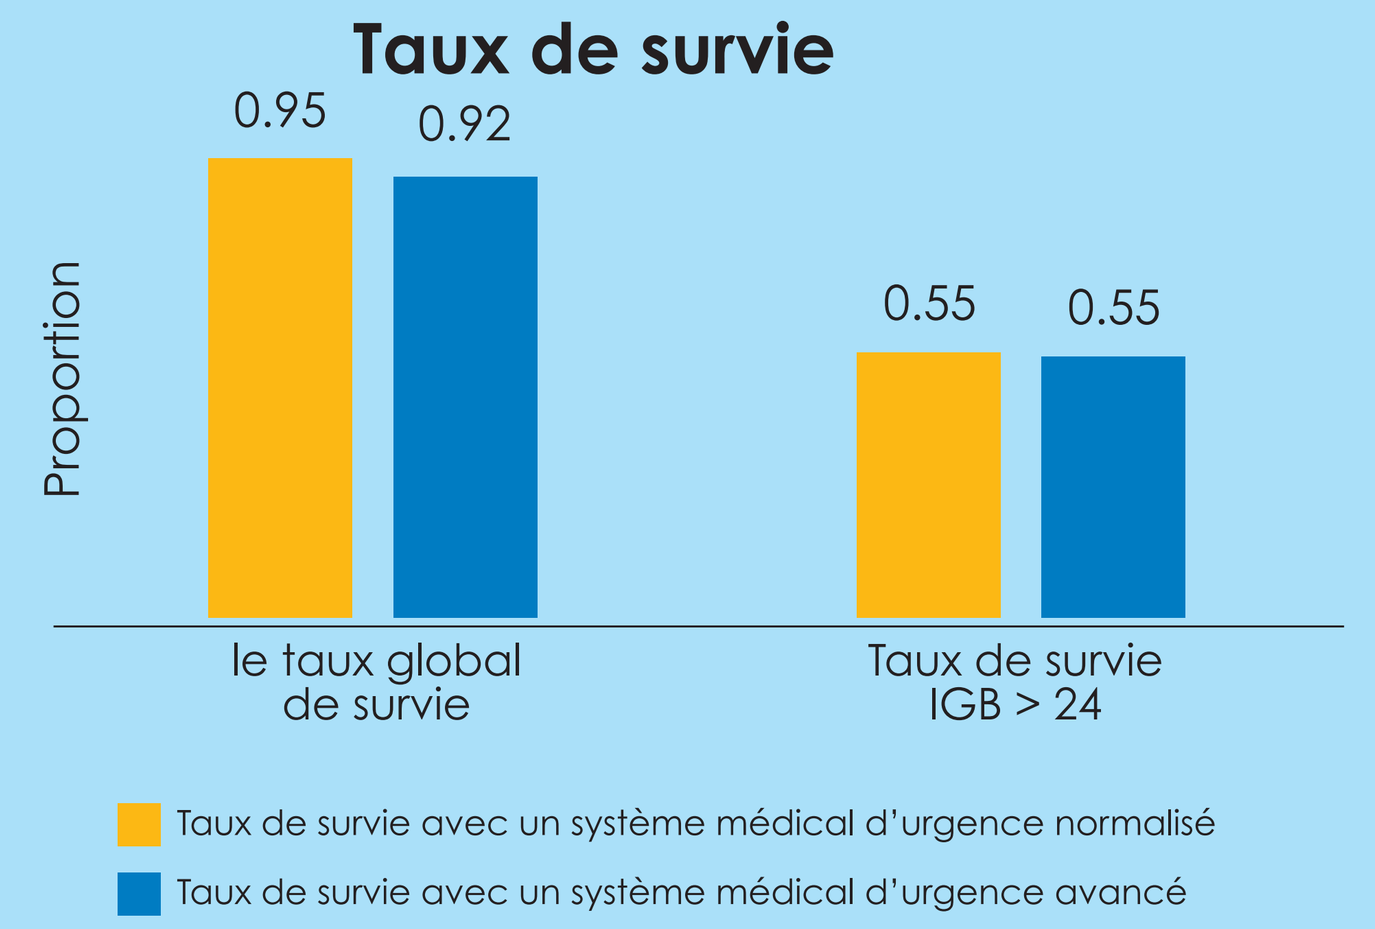

Taux de survie
Proportion
0.95
0.92
0.55
0.55
le taux global de survie
Taux de survie IGB > 24
Taux de survie avec un système médical d'urgence normalisé
Taux de survie avec un système médical d'urgence avancé

	SMUN	SMUA	Importance
Survie globale	0,95 (n = 96)	0,92 (n = 232)	*p = 0,49*
Taux de survie IGB > 24	0,55 (n = 18)	0,55 (n = 44)	*p = 1,00*
Interventions préhospitalières sur les voies respiratoires	0,03 (n = 3)	0,15 (n =38)	***p < 0,01***
Accès intraveineux préhospitalier	0,31 (n = 32)	0,78 (n = 195)	***p < 0,01***
Administration préhospitalière d'O_2	0,72 (n = 71)	0,66 (n = 165)	*p = 0,45*

Conclusion

Dans l'ensemble, le taux de survie jusqu'à l'hôpital était le même pour les systèmes médicaux d'urgence canadiens avancés et normalisés, malgré la variabilité des interventions effectuées dans chaque système. Les chiffres étant faibles, il est impossible d'exclure les avantages des cas individuels. Ces résultats soutiennent la nécessité de communiquer les données des différents cas aux autres provinces pour permettre l'analyse de variables confusionnelles potentielles.

NB Trauma Program
Programme de traumatologie du NB

Horizon Health Network
Réseau de santé Horizon

Vitalité Health Network
Réseau de santé Vitalité

Ambulance NB

New Brunswick Department of Health
Ministère de la santé du Nouveau-Brunswick